A Clinical Trial of the STAT3 Inhibitor Pyrimethamine in Chronic Lymphocytic Leukemia

Jennifer R. Brown, MD, PhD[1], Sarah R. Walker, PhD[1], Lisa N. Heppler[1], Svitlana Tyekucheva, PhD[1], Erik Nelson, PhD[1], Josephine L. Klitgaard, MSc, PhD[1], Maria Nicolais, MD[1], Yasmin Kroll[1], Michael Xiang, MD, PhD[1], Jennifer E. Yeh, MD, PhD[1], Mousumi Chaudhury[1], Zachary T. Giaccone[1], Stacey Fernandes[1], Jeffrey G. Supko, PhD[2], Catherine J. Wu, MD[1] and David A. Frank, MD, PhD[1]

[1]Dana-Farber Cancer Institute, Boston and [2]Massachusetts General Hospital, Boston

Background

- Despite advances in targeted therapy, chronic lymphocytic leukemia remains a highly prevalent and essentially incurable malignancy.
- One molecular hallmark of CLL is the constitutive serine phosphorylation and transcriptional activation of the oncogenic transcription factor STAT3.
- Targeting STAT3 may have a high therapeutic index since normal cells can tolerate loss of STAT3 function.

Objective

- To determine whether STAT3 inhibition would confer clinical benefit in patients with CLL, we conducted a phase I clinical trial of continuous daily pyrimethamine, in the era before targeted therapy (enrollment 5/2010 – 4/2012), in relapsed CLL patients whose disease progressed despite standard therapies. ClinicalTrials.gov Identifier: NCT01066663

Methods and Study Design

- We screened a chemical library of drugs known to be safe in humans for specific inhibitors of STAT3-dependent transcription to identify STAT3 inhibitors that could be rapidly introduced into proof-of-concept clinical trials.
- Based on the identification of pyrimethamine as a specific STAT3 inhibitor, we used a 3+3 dose escalation design with three cohorts: 12.5, 25, and 50 mg per day (mg/d). Eligibility included relapsed refractory CLL with progressive disease in need of therapy, without any threshold values of neutrophils, hemoglobin or platelets.
- The primary endpoint of the trial was to determine the maximum tolerated and recommended phase 2 dose of pyrimethamine in CLL patients. The DLT evaluation period was 28 days. DLT was defined as any grade 3 or higher non-hematologic toxicity that was not rapidly remediable with supportive care, with only grade 4 infections considered dose-limiting. Hematologic toxicity was graded by iwCLL criteria.
- We defined a signature of STAT3-dependent genes that are upregulated in CLL cells relative to normal B lymphocytes and then quantitated expression of these genes from the patients' CLL cells at baseline and while on pyrimethamine to determine whether pyrimethamine was inhibiting STAT3-dependent gene expression *in vivo*.
- We measured trough concentrations of pyrimethamine in plasma and in peripheral blood mononuclear cells at distinct time points for patients on the trial to determine whether the levels of pyrimethamine achieved in patients corresponded with levels known to inhibit STAT3 transcriptional function *ex vivo*.
- Samples for PK/PD analysis were drawn weekly in the first month and every other week in the second month on therapy.
- We also quantitated the cytotoxic effect of pyrimethamine on the pre-treatment cells of patients *ex vivo* to generate predictors of response.

Pyrimethamine Inhibits STAT3 Transcriptional Activity and Decreases Viability of CLL Cells

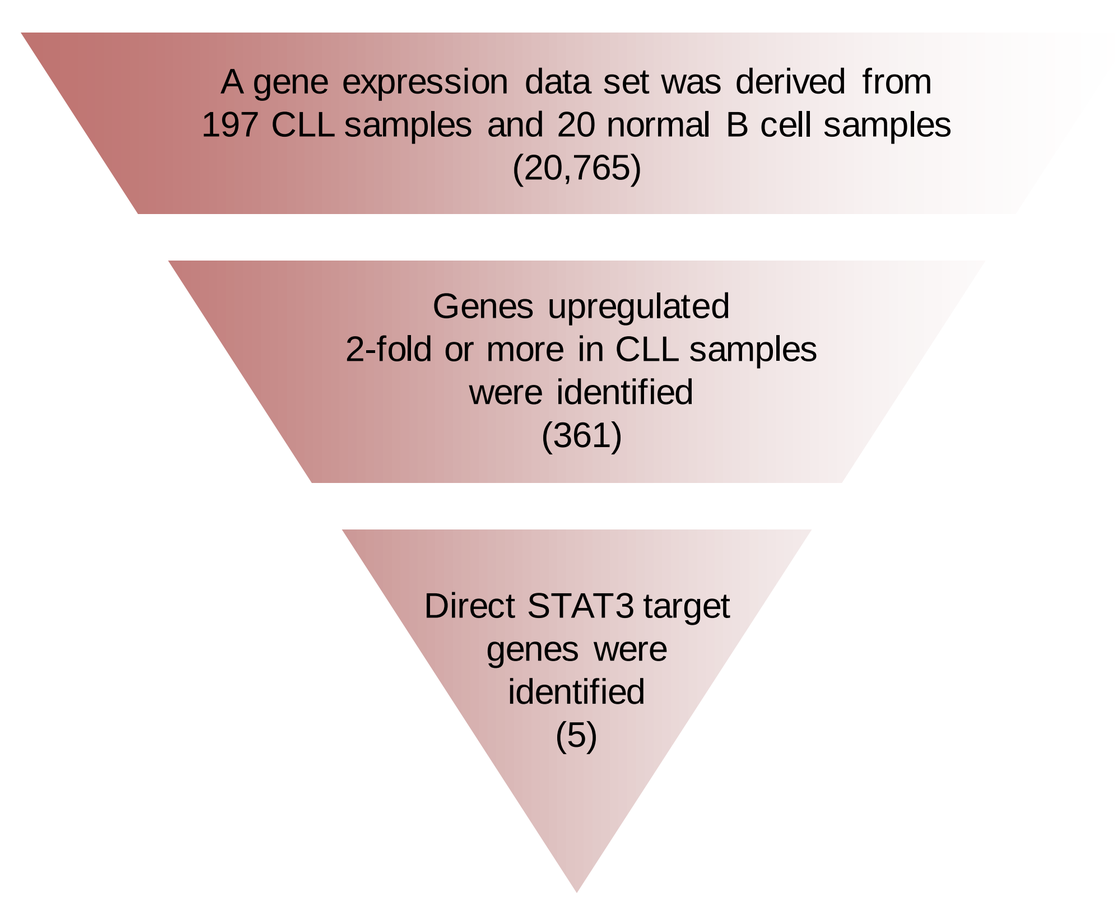
A gene expression data set was derived from 197 CLL samples and 20 normal B cell samples (20,765)
Genes upregulated 2-fold or more in CLL samples were identified (361)
Direct STAT3 target genes were identified (5)

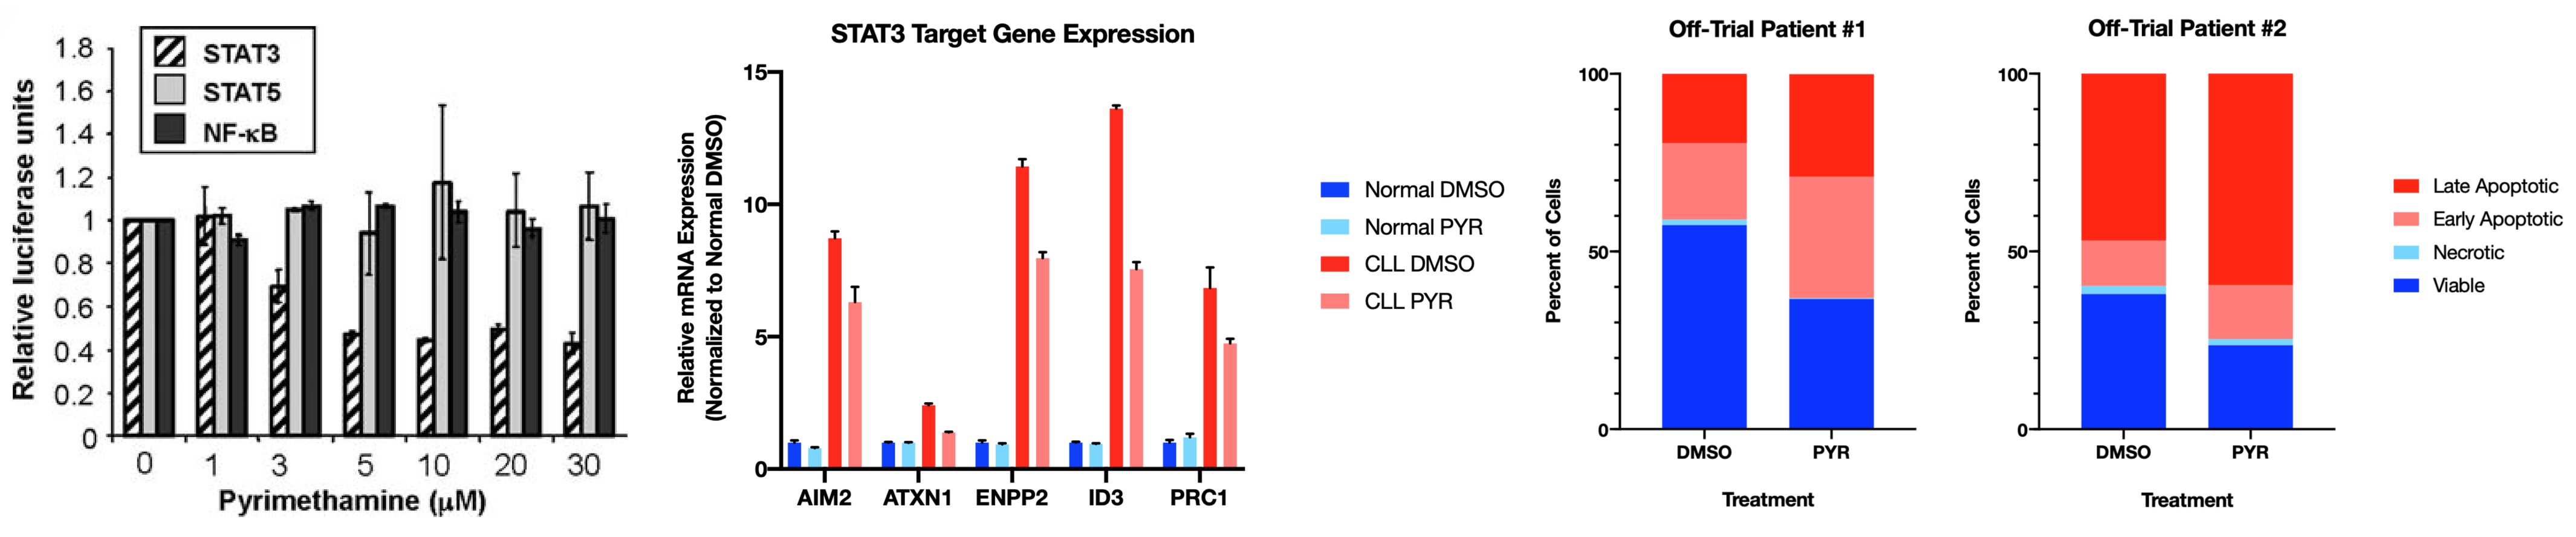
STAT3
STAT5
NF-κB
Relative luciferase units
Pyrimethamine (μM)
STAT3 Target Gene Expression
Relative mRNA Expression (Normalized to Normal DMSO)
AIM2
ATXN1
ENPP2
ID3
PRC1
Normal DMSO
Normal PYR
CLL DMSO
CLL PYR
Off-Trial Patient #1
Off-Trial Patient #2
Percent of Cells
DMSO
PYR
Treatment
Late Apoptotic
Early Apoptotic
Necrotic
Viable

Human Molecular Genetics, 2011, Vol. 20, No. 21

Identification of STAT3-regulated genes in CLL.

Pyrimethamine was identified from a chemical library screen of inhibitors of STAT3 transcriptional function. Pyrimethamine inhibits STAT3-dependent gene expression and induces apoptosis of CLL cells treated *in vitro*.

Patient Characteristics and Clinical Response

N = 16	N (range or %)
Age at Study Entry	63 (36-85)
Sex - Male	11 (69)
Rai Stage 3-4	4 (25)
Median WBC at Baseline	11 (2 - 94.9)
Median Hemoglobin at Baseline	10 (7.6 - 15)
Median Platelets at Baseline	113 (6 - 217)
Median β2m	5 (3 - 16.5)
ECOG 0 / 1 / 2	3 (19) / 10 (62) / 2 (13)
Time from Diagnosis to Study Therapy	74 months (6-176)
Number of Prior Therapies	6 (1-16)
IGHV Unmutated	11 / 14 (79%)
ZAP-70 Positive (>20%)	8 / 9 (89%)
Interphase FISH (Dohner hierarchy)	
Del 17p	6 (38%)
Del 11q	3 (19%)
Trisomy 12	4 (25%)
Normal	2 (13%)
Del 13q	1 (6%)

- Sixteen heavily pretreated patients enrolled in phase I. Three patients each enrolled on cohorts 1 and 2, and ten patients enrolled on cohort 3. No DLTs were observed, but four patients on cohort 3 came off study before completing the 28 day DLT evaluation period (three for progressive disease and one with pneumonia), and were replaced.
- Most observed toxicity was clearly disease-related. Notable toxicities that were possibly drug related included asymptomatic reversible grade 3 transaminitis, and two cases of transient grade 4 thrombocytopenia.
- No objective responses by iwCLL criteria were observed. 50% of patients achieved stable disease, with one patient dosed at 50 mg/d on therapy for 12 months, and two at 25 mg/d remained on for 4 and 6 months. The remaining patients had progressive disease, with all but one patient discontinuing for progressive disease. The median time on therapy was 1.5 months (0.23-9.99).
- Clinically the MTD was not reached.

Pharmacokinetic Analyses

Patient Group	Plasma Concentration (μM)	Intracellular Concentration (pg/10^6 cells)
Cohort 1 (12.5 mg/day; n=3)	1.4 (Range: 1.1 – 1.8)	116.4
Cohort 2 (25 mg/day; n=3)	2.6 (Range: 2.1 – 3.0)	338.8
Cohort 3 (50 mg/day; n=10)	5.7 (Range: 2.5 – 8.3)	724.1

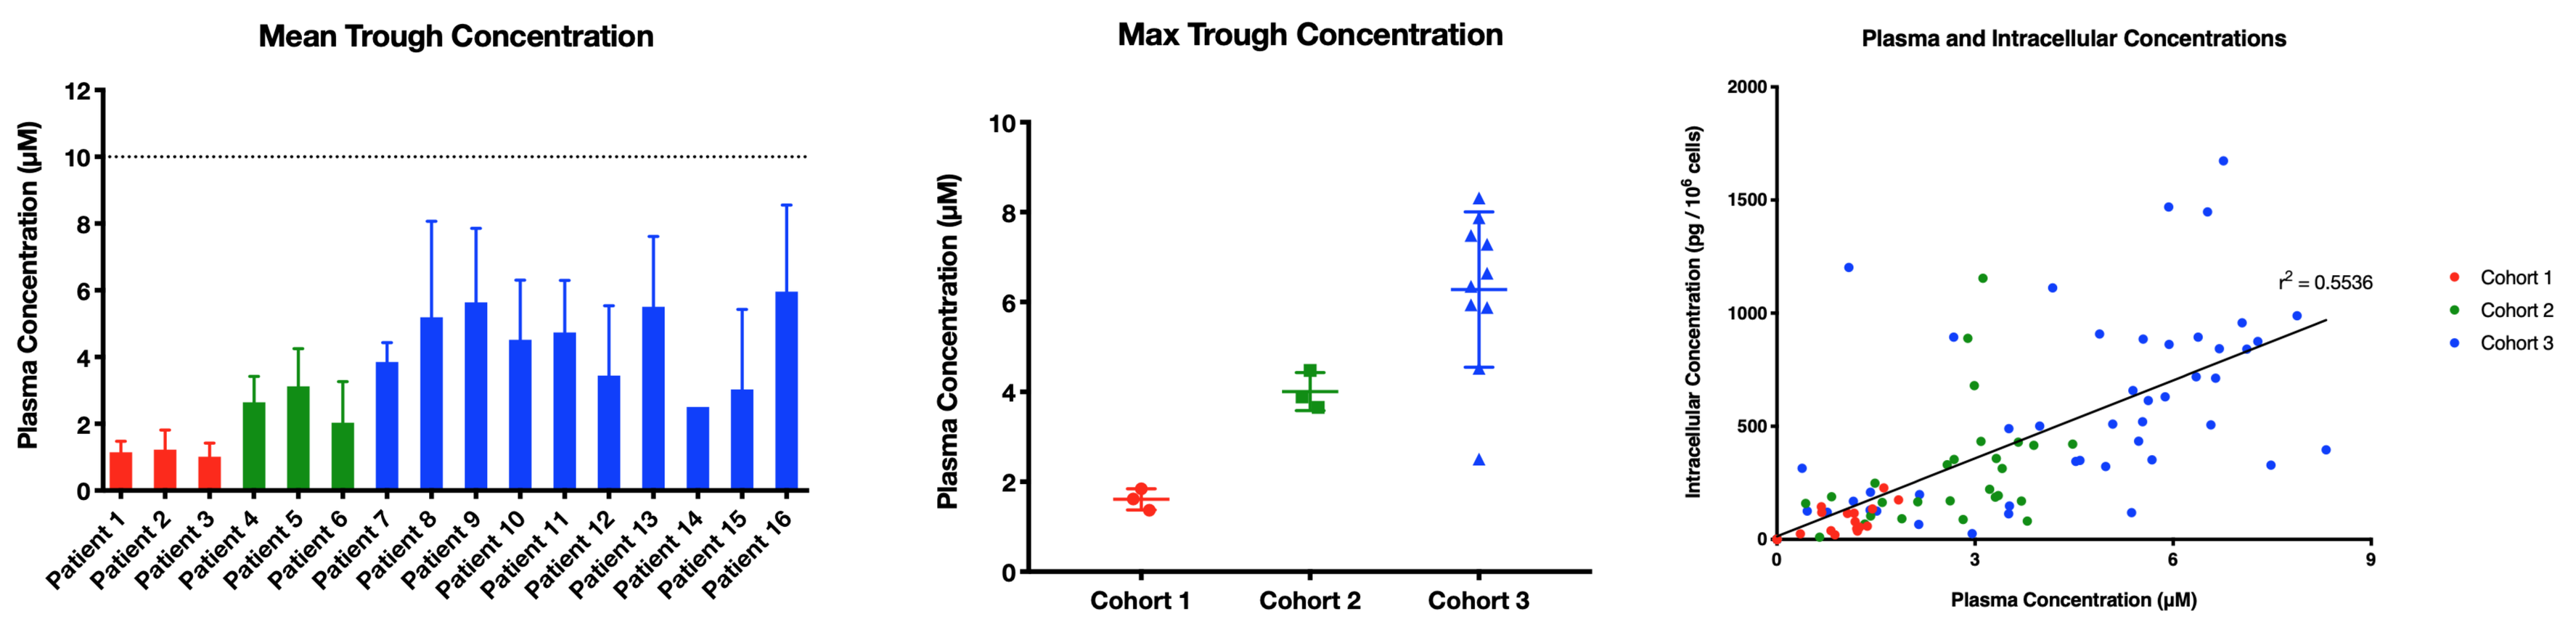
Mean Trough Concentration
Plasma Concentration (μM)
Max Trough Concentration
Cohort 1
Cohort 2
Cohort 3
Plasma and Intracellular Concentrations
Intracellular Concentration (pg / 10^6 cells)
Plasma Concentration (μM)
r² = 0.5536

Mean trough pyrimethamine concentrations. (Determined on day 15 of the first cycle, or closest blood draw following day 15).

Levels of pyrimethamine achieved in patients increased with dose, and intracellular concentration correlated with plasma concentration. However, plasma concentrations were generally below the level of 8 to 10 μM necessary for maximal inhibition of STAT3 transcriptional activity.

Pharmacodynamic Analyses

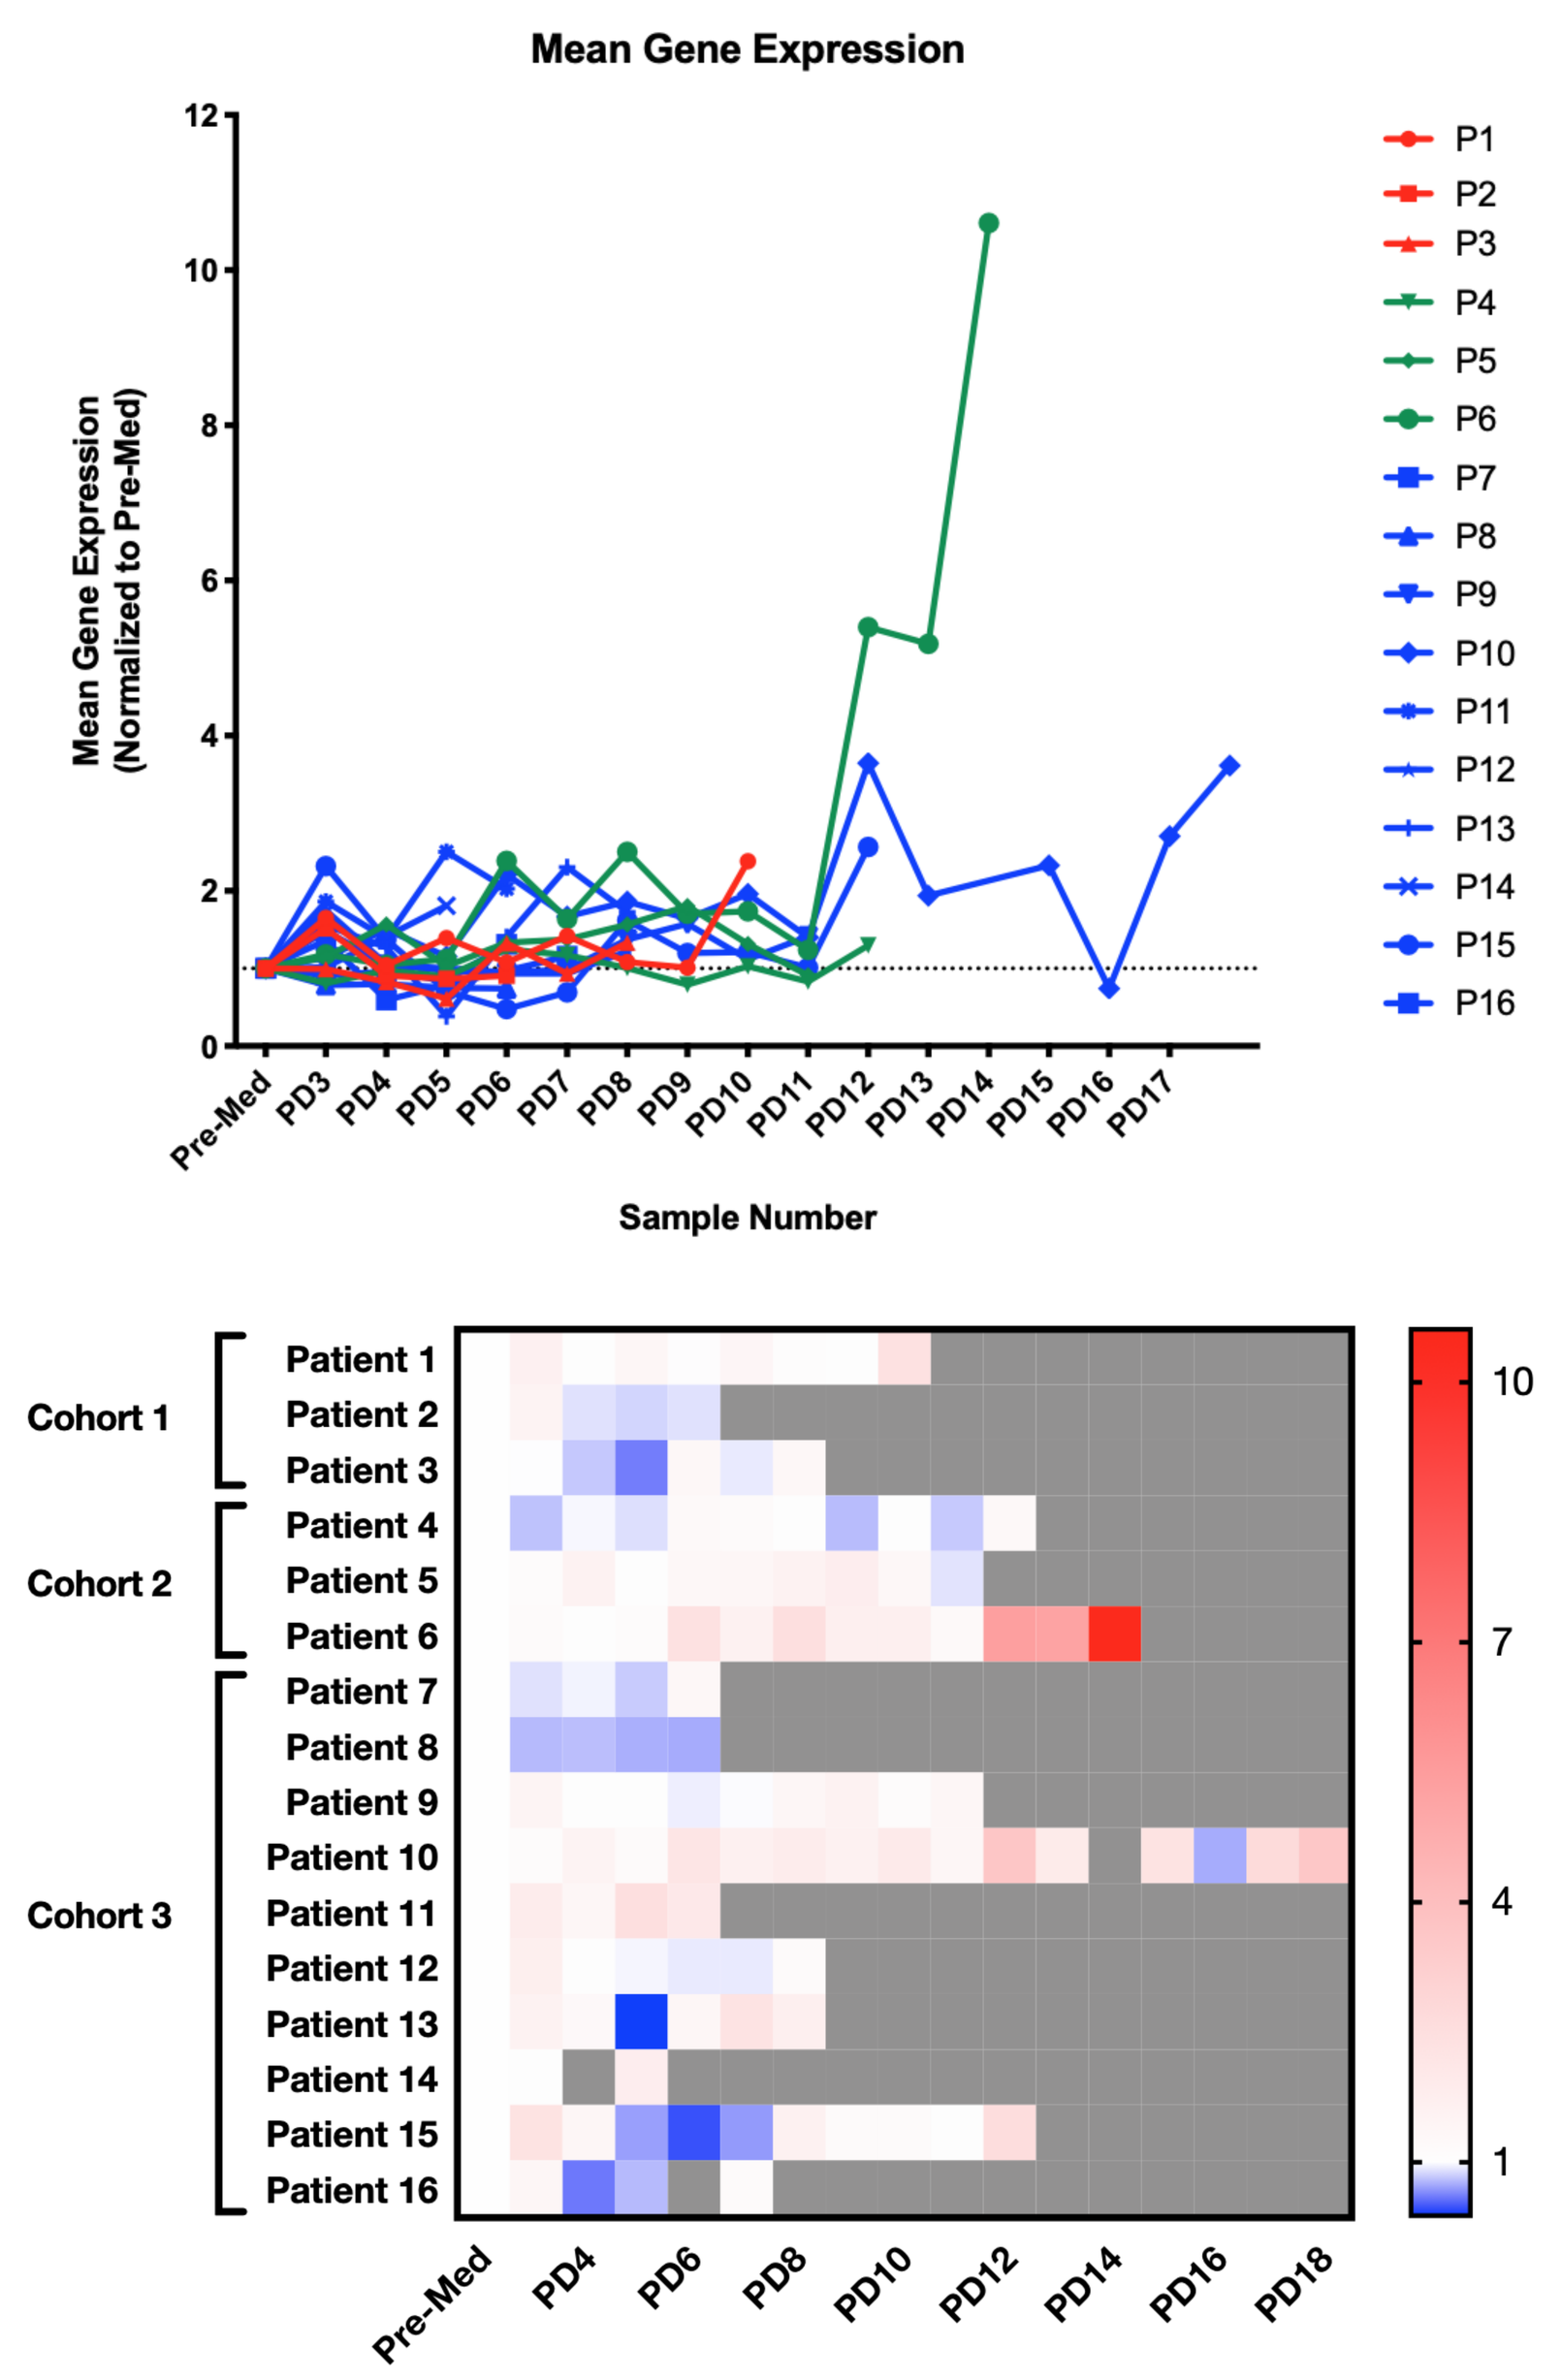
Mean Gene Expression
Mean Gene Expression (Normalized to Pre-Med)
Sample Number
Cohort 1
Cohort 2
Cohort 3

The response of STAT3 target genes *in* vivo while on pyrimethamine was variable. The mean expression of the five STAT3 target genes was measured from patient samples taken at distinct time points to determine whether pyrimethamine was inhibiting STAT3 activity *in vivo.* PD samples were drawn weekly in the first month and every 2-4 weeks on therapy thereafter.

Conclusions

- Pyrimethamine is an inhibitor of STAT3 transcriptional function that decreases survival of CLL cells and can safely be given to patients with CLL.
- Given that PK and PD data suggest that adequate drug levels for target inhibition were not achieved, it may be necessary to increase the daily dose of pyrimethamine to beyond 50 mg/day to adequately determine whether this drug can exert an on-target therapeutic effect in CLL patients.
- Since STAT3 target genes include many immune-suppressive and anti-apoptotic genes, it is likely that pyrimethamine will be most beneficial when combined with other therapeutic modalities.

Acknowledgement

- This clinical trial was supported by a grant from the Lymphoma Research Foundation.

Disclosures: Brown: *Pharmacyclics*: Consultancy; *Celgene*: Consultancy; *Abbvie*: Consultancy; *Boehringer*: Consultancy; *Sun Pharmaceutical Industries*: Research Funding; *Gilead*: Consultancy, Research Funding; *Loxo*: Consultancy; *Verastem*: Consultancy, Research Funding; *Invectys*: Membership on an entity's Board of Directors or advisory committees; *Morphosys*: Membership on an entity's Board of Directors or advisory committees; *Roche/Genentech*: Consultancy; *Janssen*: Consultancy; *Acerta / Astra-Zeneca*: Membership on an entity's Board of Directors or advisory committees; *Sunesis*: Consultancy; *Genentech*: Consultancy; *Beigene*: Membership on an entity's Board of Directors or advisory committees; *TG Therapeutics*: Consultancy. **Wu:** *Neon Therapeutics*: Equity Ownership.